Combining OP-3136, a KAT6 Inhibitor, With Endocrine Therapy and CDK4/6 Inhibitor Enhances Anti-tumor Activity in ER+/HER2- Breast Cancer Models

Abstract # 230

Gopinath S. Palanisamy[1], Susanna A. Barratt[1], Chelsea Hope[1], Brandon Robello[1], Guadalupe Peña[1], Megha Goyal[2], Chandregowda Venkateshappa[2], Raymond A. Ng[1], Murali Ramachandra[2], David C. Myles[1]

[1]Olema Oncology, San Francisco, CA; [2]Aurigene Oncology, Bangalore, India

Background

- The histone lysine acetyl transferases, KAT6A and KAT6B, are enzymes involved in epigenetic regulation of oncogenes[1]
- KAT6 enzymes acetylate histone H3 which unwinds the chromatin resulting in gene transcription. Inhibition of KAT6 stops acetylation and hinders breast cancer cell proliferation through downregulation of genes involved in estrogen signaling and other signaling pathways[2] (**Figure 1**)
- Dysregulation of KAT6 activity (via gene amplification, overexpression, fusion, mutation, etc.) is observed in many cancers, including breast cancer[1,3]

Figure 1: Inhibition of KAT6 enzymes blocks histone acetylation and downregulates transcription of proliferation-associated genes

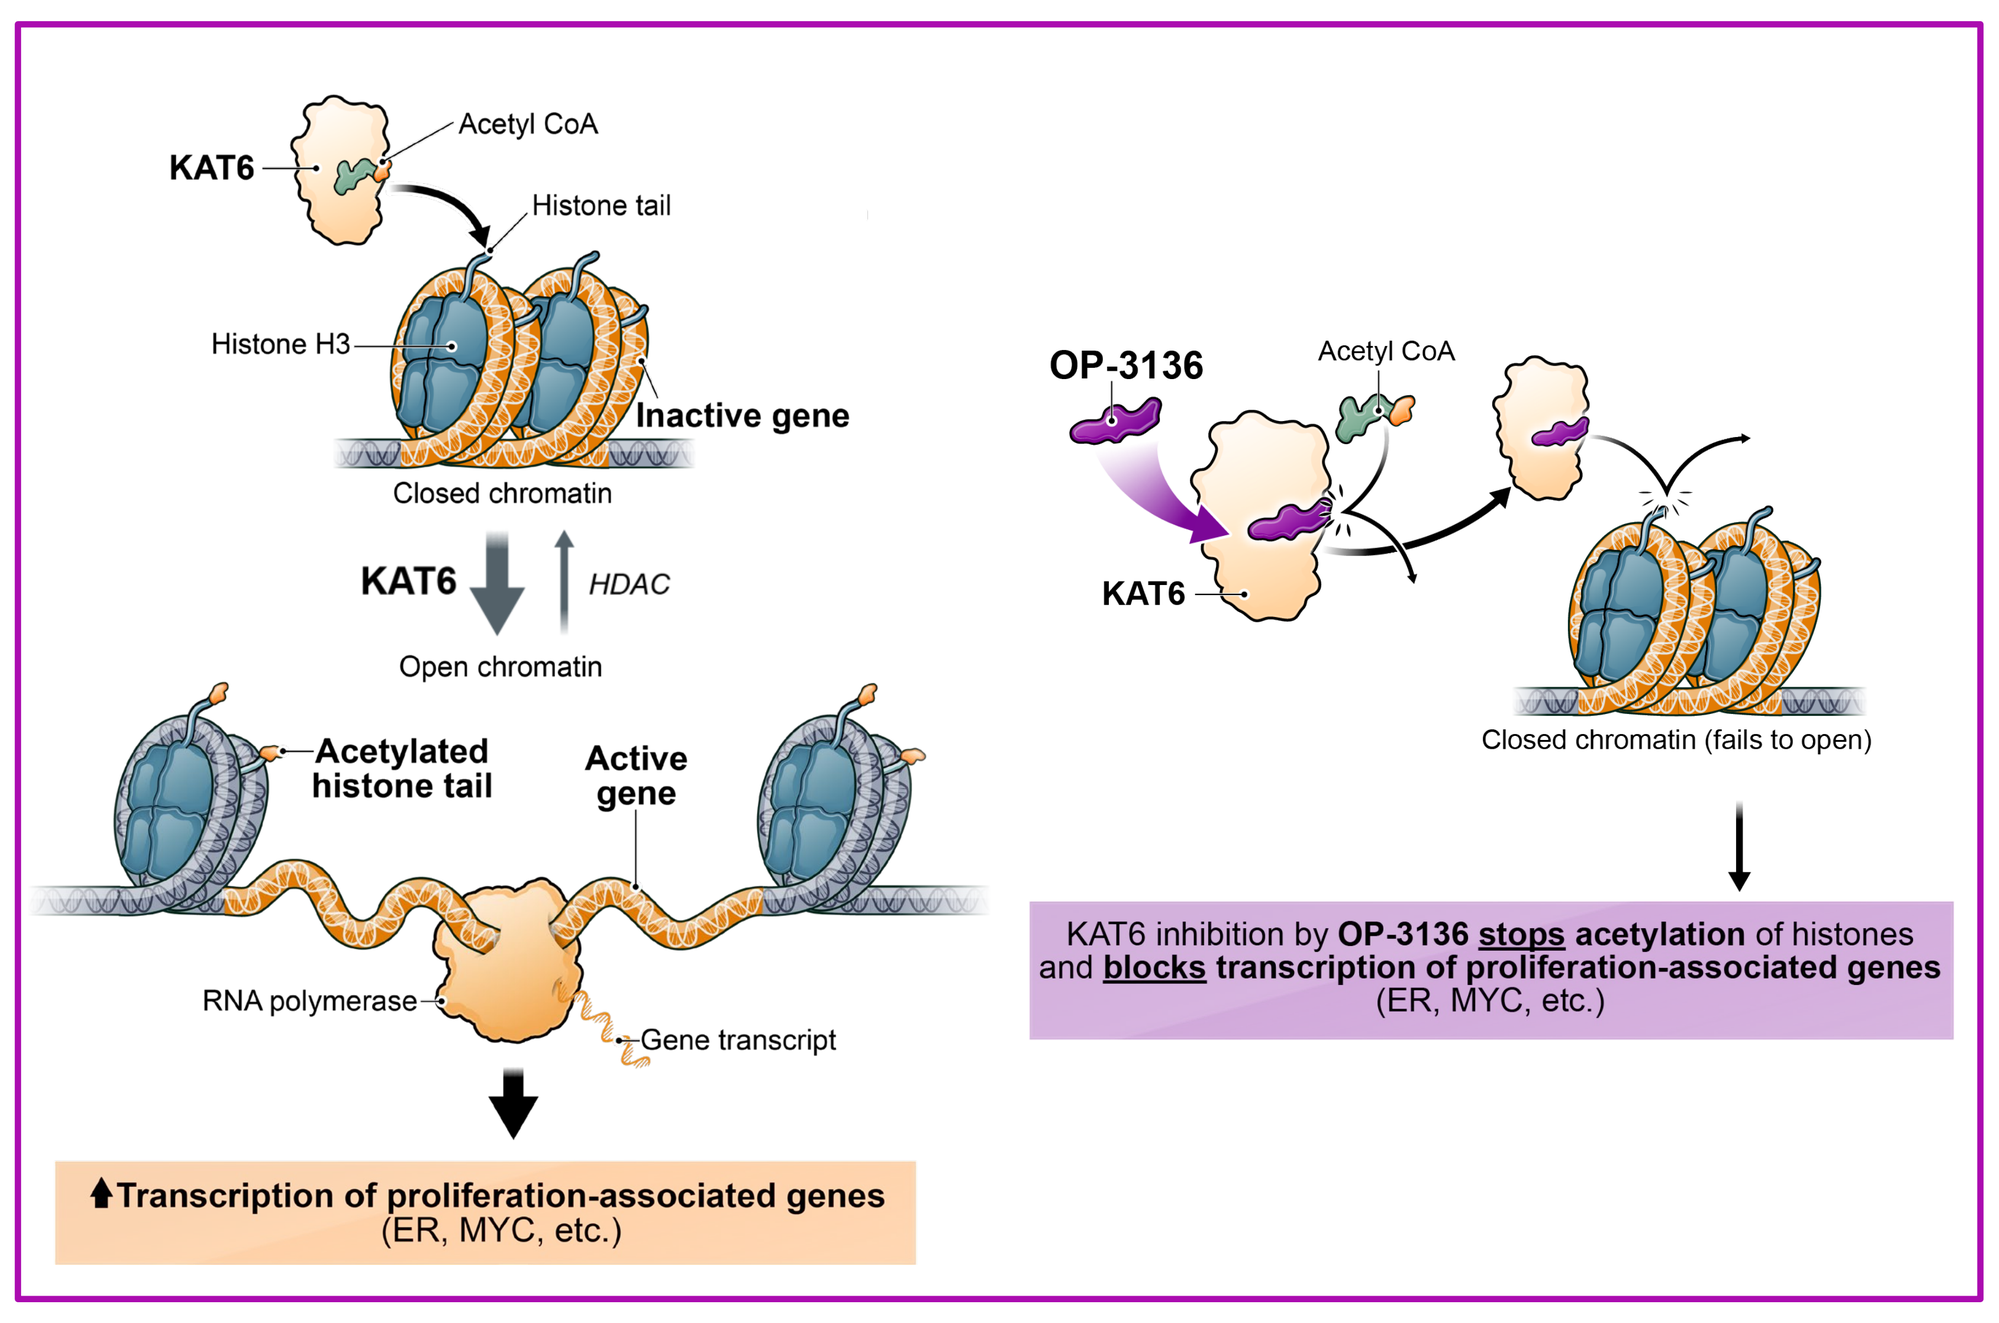
KAT6
Acetyl CoA
Histone tail
Histone H3
Inactive gene
Closed chromatin
KAT6
HDAC
Open chromatin
Acetylated histone tail
Active gene
RNA polymerase
Gene transcript
Transcription of proliferation-associated genes (ER, MYC, etc.)
OP-3136
Acetyl CoA
KAT6
Closed chromatin (fails to open)
KAT6 inhibition by OP-3136 stops acetylation of histones and blocks transcription of proliferation-associated genes (ER, MYC, etc.)

CoA, coenzyme A; **ER**, estrogen receptor; **HDAC**, histone deacetylase; **H3,** histone 3**; KAT**, lysine acetyltransferase; **MYC,** MYC proto-oncogene; **RNA**, ribonucleic acid.

- KAT6 inhibitors block ERα and cyclin D1 (*CCND1*) genes at the transcriptional level, and combining them with anti-estrogens and cyclin-dependent kinase 4/6 (CDK4/6) inhibitors that act at the ligand level may result in enhanced anti-proliferative activity (**Figure 2**)[2]
- Combination of a KAT6 inhibitor with fulvestrant has shown promise in a Phase 1 clinical trial enrolling patients with estrogen receptor-positive (ER+)/human epidermal growth factor receptor 2-negative (HER2-) metastatic breast cancer[4]
- OP-3136 is a highly selective KAT6 inhibitor in preclinical development that has been shown to have significant anti-proliferative activity in ER+ breast cancer models[5]

Figure 2: Combination of KAT6 inhibitors with anti-estrogens and CDK4/6 inhibitors may enhance blockade of ER signaling and cell cycle progression

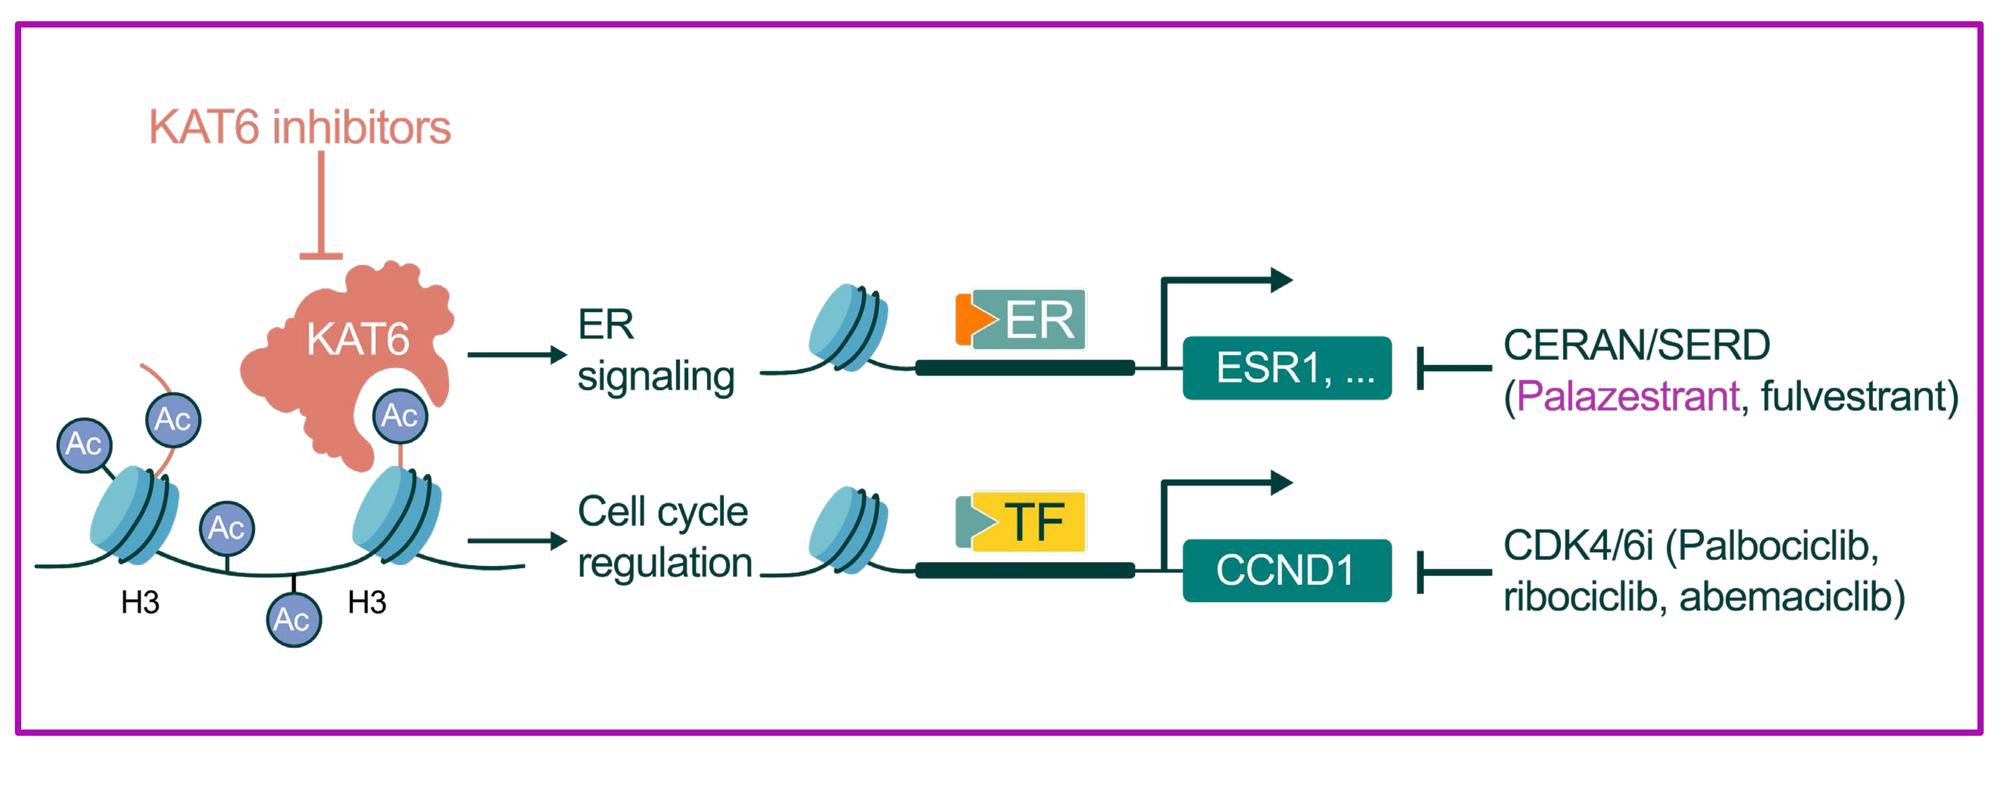
KAT6 inhibitors
KAT6
Ac
H3
ER signaling
ER
ESR1, ...
CERAN/SERD (Palazestrant, fulvestrant)
Cell cycle regulation
TF
CCND1
CDK4/6i (Palbociclib, ribociclib, abemaciclib)

Ac, acetyl group; **CCND1,** cyclin D1; **CDK4/6i,** cyclin-dependent kinase 4/6 inhibitor; **CERAN,** complete estrogen receptor antagonist; **ER,** estrogen receptor; **ESR1,** estrogen receptor 1; **H3,** histone H3; **KAT,** lysine acetyltransferase; **SERD,** selective estrogen receptor degrader; **TF,** transcription factor.

Methods

- **Cell proliferation assays:** T47D cells were plated in 96-well plates at 1000 cells/well in complete medium and incubated overnight. Cells were treated with serial dilutions of OP-3136 plus fixed concentrations of fulvestrant, palazestrant, and/or ribociclib in the presence of 100 pM estradiol for 10 days. Cell number was assessed using CellTiter-Glo 2.0 and normalized to T=0.
- **Drug combination analysis:** Synergy was evaluated using SynergyFinder 3.0 tools, specifically the Zero Interaction Potency (ZIP) model, which assumes that the combined effect of two molecules is the sum of their individual responses without any interaction. Combinations were normalized to the monotherapy response; deviations >10 from this predicted combined effect indicate synergy (greater effect), between -10 and +10 indicate additive (equal) effect, and <10 indicate antagonism (lesser effect).
- **Xenograft studies:** Athymic, female nude mice (immune deficient) were supplemented with estradiol and implanted subcutaneously with an ER+ breast cancer cell line, T47D, or a patient-derived xenograft tumor fragment in the mammary fat region or flank. Mice were randomized into groups when the tumor volume reached ~150 mm^3 and were treated for at least 28 days with either vehicle or the indicated regimens.
- **Comparator compound**: PF compound E from the published patent[6] was used as a positive control comparator.

Results

OP-3136 synergizes with anti-estrogens and CDK4/6 inhibitors in vitro to inhibit proliferation of an *ESR1*^{WT} ER+ breast cancer cell line

Figure 3: Dose-response curves (**A-C**) and synergy plots (**D-F**) for doublet combinations in T47D cells

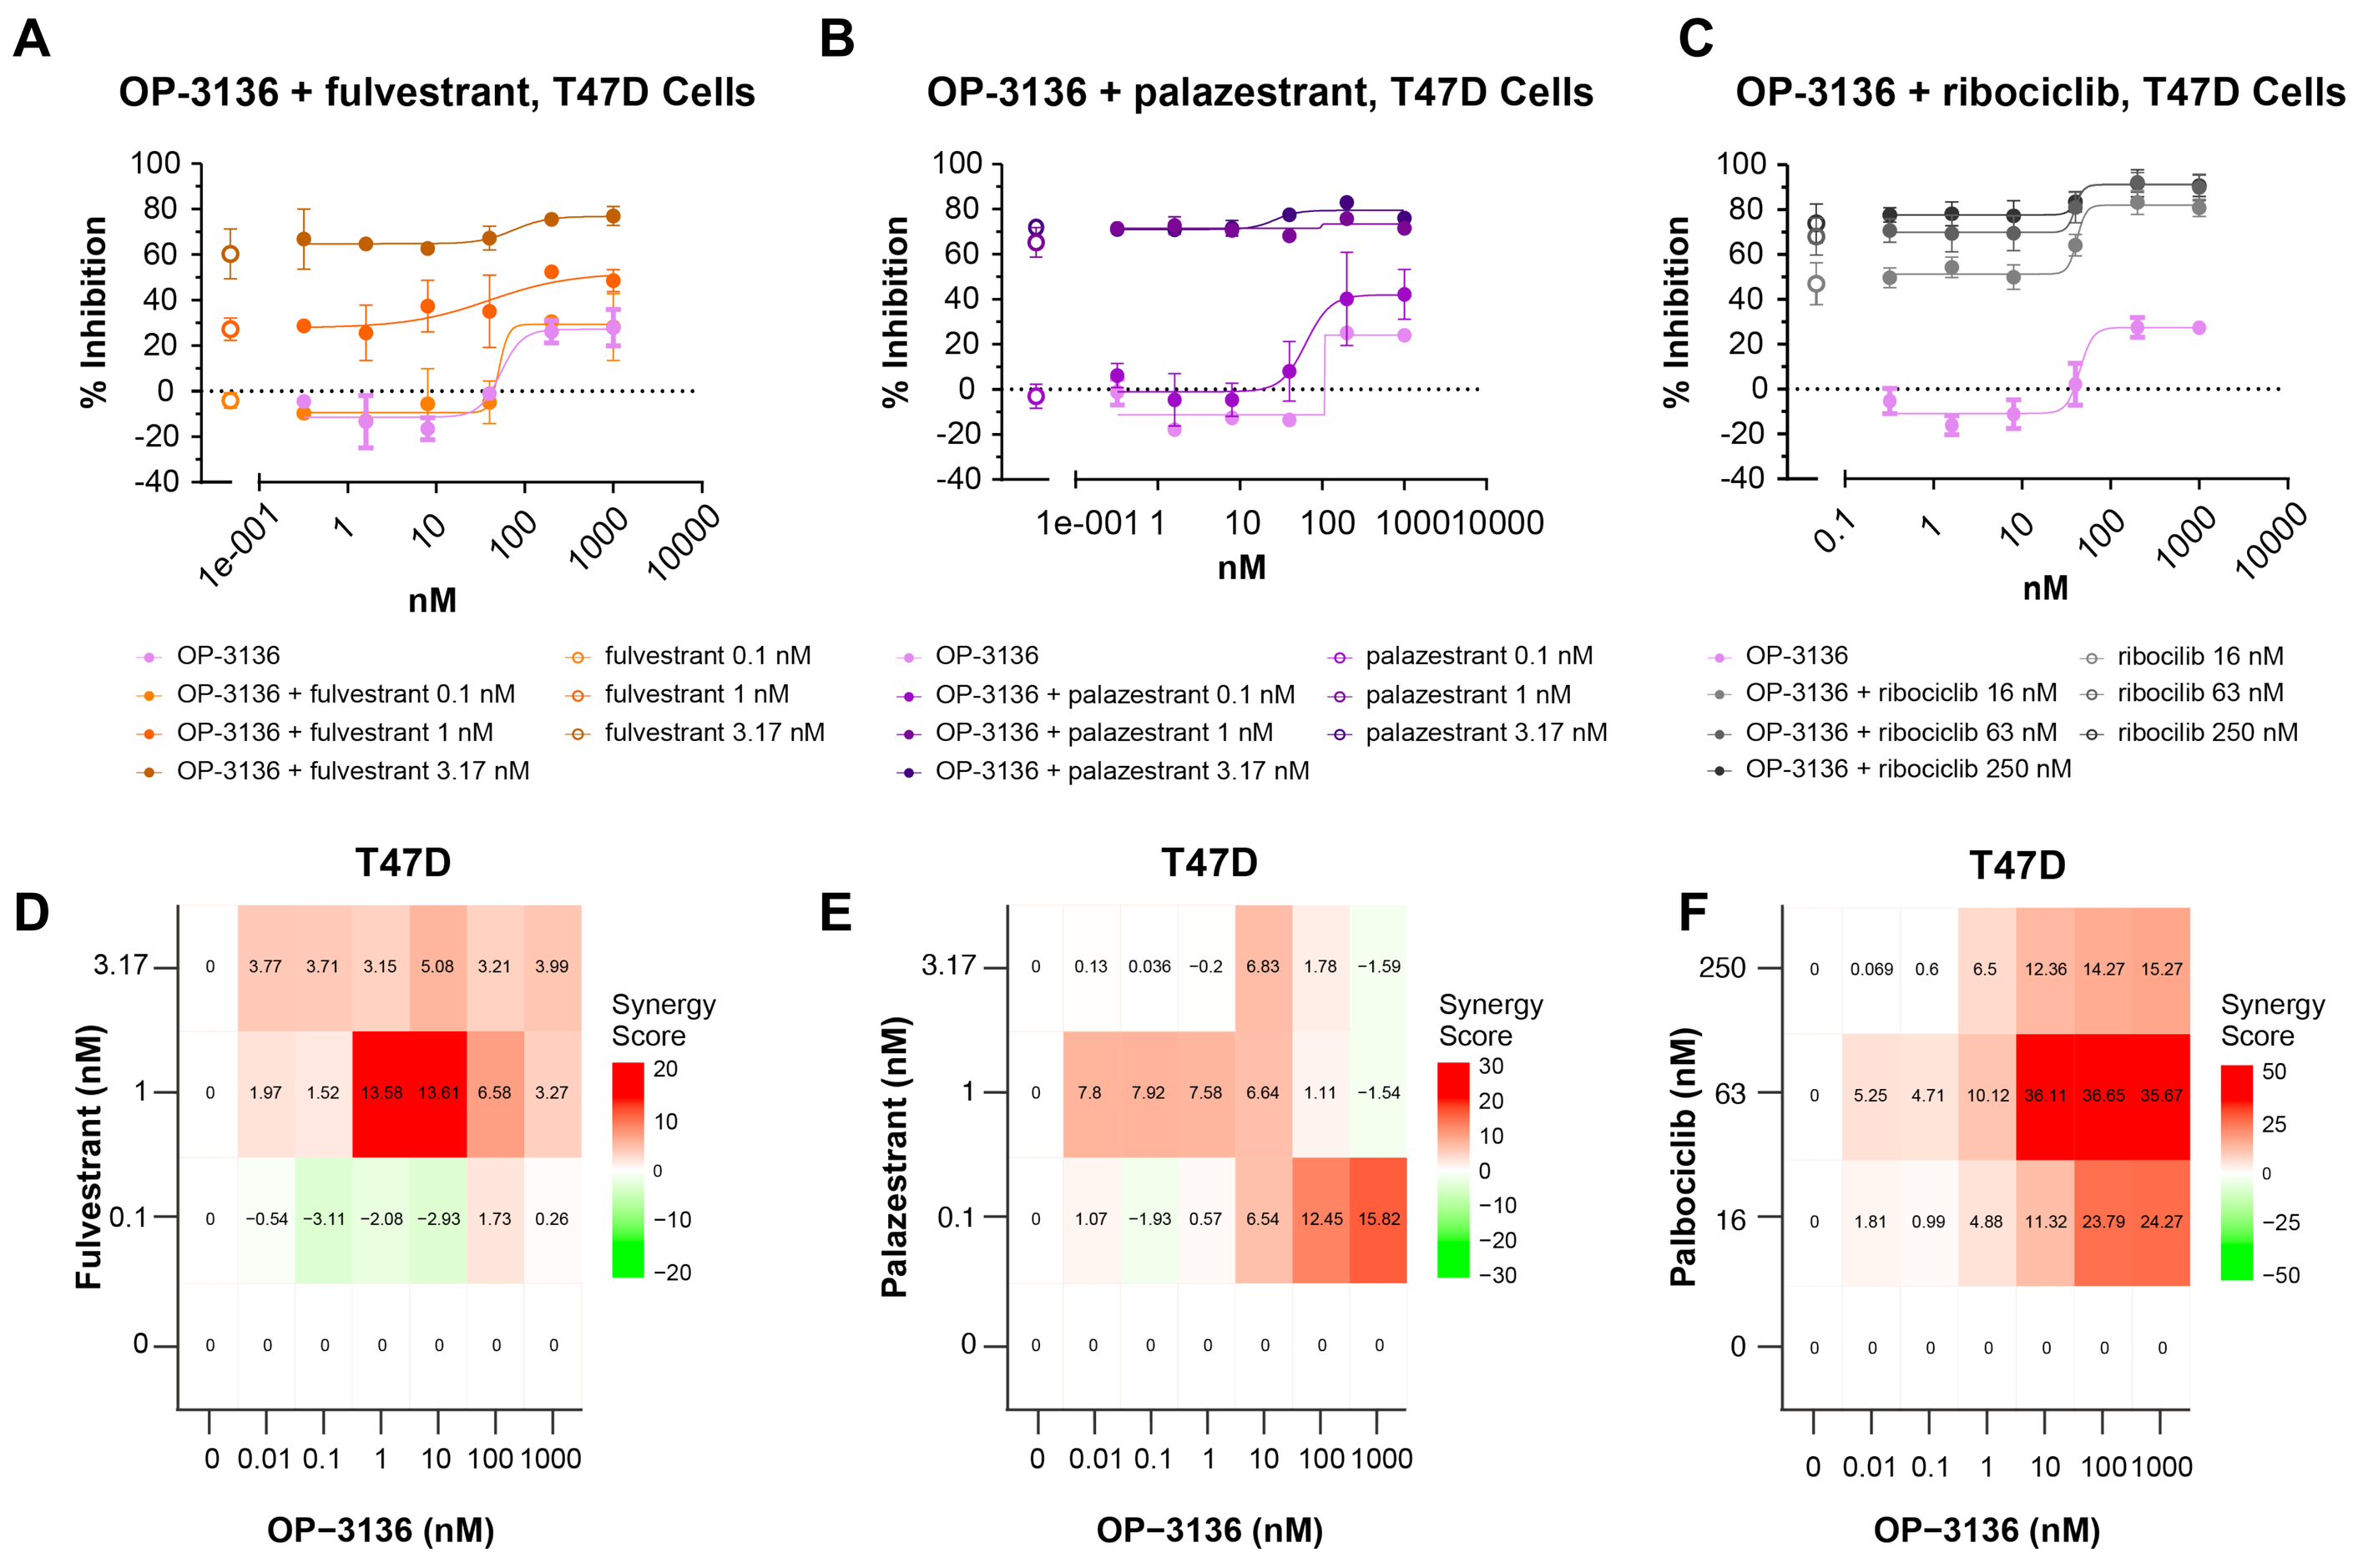
A
OP-3136 + fulvestrant, T47D Cells
% Inhibition
nM
OP-3136
OP-3136 + fulvestrant 0.1 nM
OP-3136 + fulvestrant 1 nM
OP-3136 + fulvestrant 3.17 nM
fulvestrant 0.1 nM
fulvestrant 1 nM
fulvestrant 3.17 nM
B
OP-3136 + palazestrant, T47D Cells
OP-3136 + palazestrant 0.1 nM
OP-3136 + palazestrant 1 nM
OP-3136 + palazestrant 3.17 nM
palazestrant 0.1 nM
palazestrant 1 nM
palazestrant 3.17 nM
C
OP-3136 + ribociclib, T47D Cells
OP-3136 + ribociclib 16 nM
OP-3136 + ribociclib 63 nM
OP-3136 + ribociclib 250 nM
ribociclib 16 nM
ribociclib 63 nM
ribociclib 250 nM
D
T47D
Fulvestrant (nM)
OP−3136 (nM)
Synergy Score
E
Palazestrant (nM)
F
Palbociclib (nM)

Figure 4: Dose-response curves for triplet combinations with fulvestrant (**A**) and palazestrant (**B**) in T47D cells

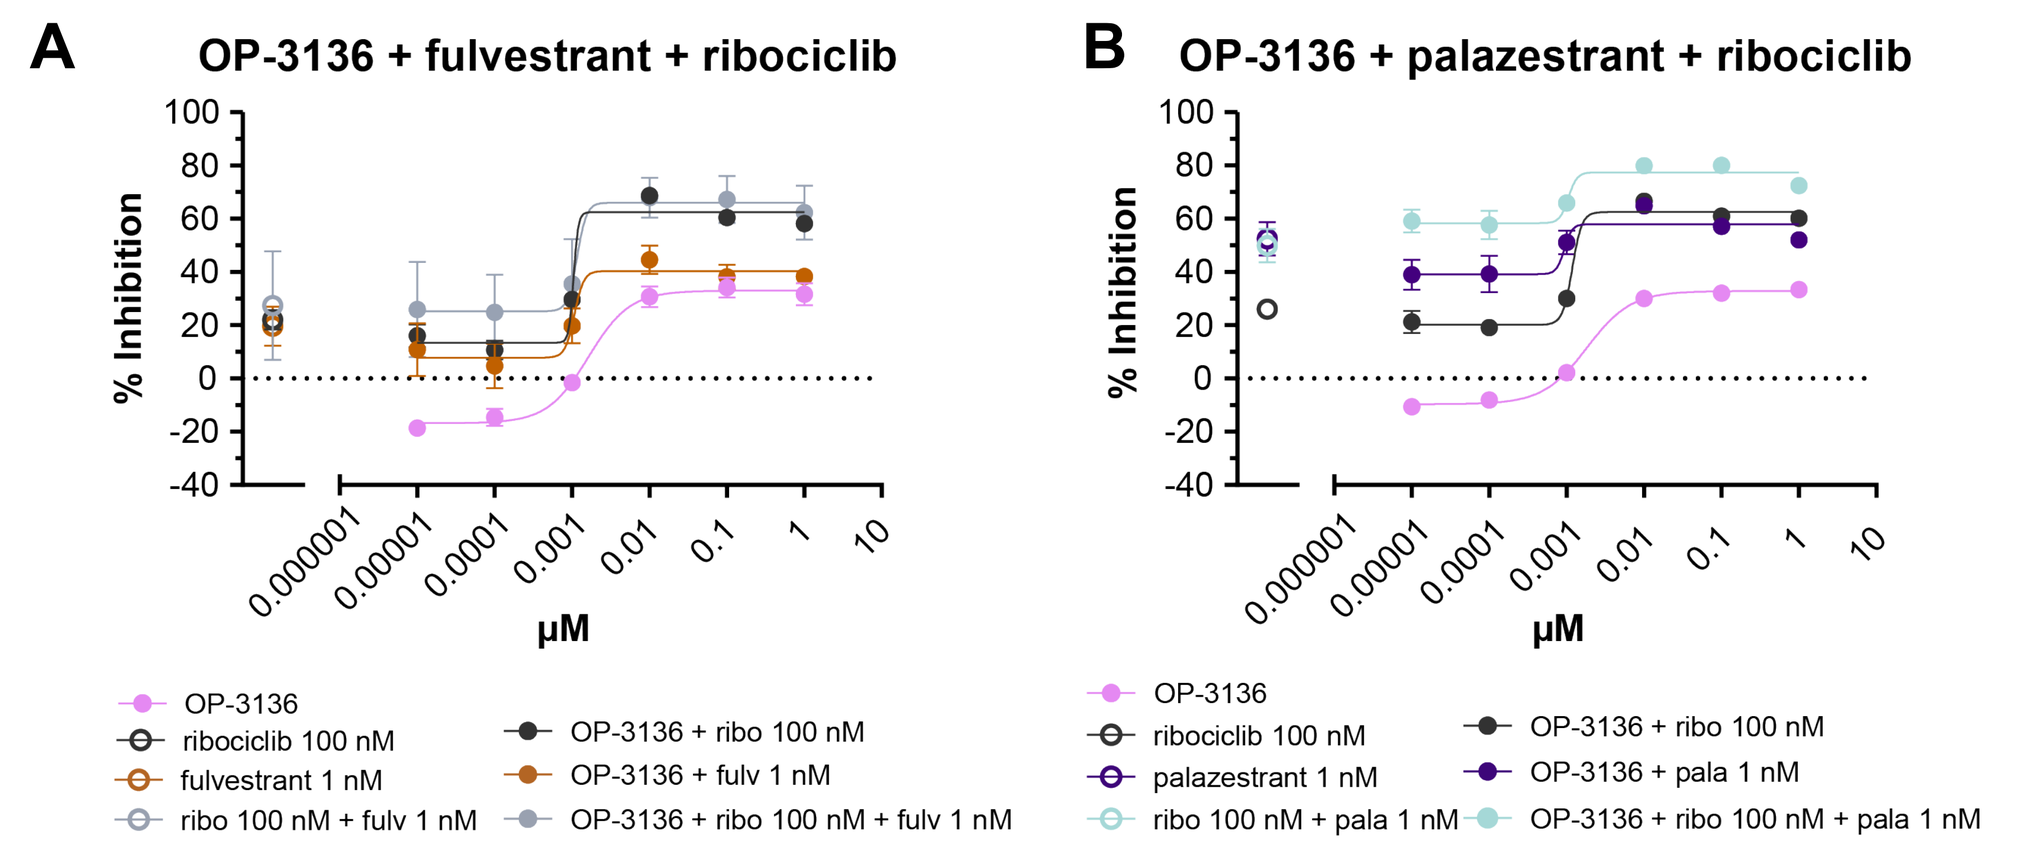
A
OP-3136 + fulvestrant + ribociclib
% Inhibition
μM
OP-3136
ribociclib 100 nM
fulvestrant 1 nM
ribo 100 nM + fulv 1 nM
OP-3136 + ribo 100 nM
OP-3136 + fulv 1 nM
OP-3136 + ribo 100 nM + fulv 1 nM
B
OP-3136 + palazestrant + ribociclib
palazestrant 1 nM
ribo 100 nM + pala 1 nM
OP-3136 + pala 1 nM
OP-3136 + ribo 100 nM + pala 1 nM

- Combination of OP-3136 with fulvestrant, palazestrant, or ribociclib (**Figure 3A-C**) enhanced inhibition of ER+/HER2-, *ESR1*^{WT} T47D cells over monotherapy
- Synergy analysis indicated that OP-3136 acts synergistically when combined with anti-estrogens or CDK4/6 inhibitors (**Figure 3D-F**)
- Combining OP-3136 with both fulvestrant and ribociclib (triplet combination) did not demonstrate additional anti-proliferative effects compared to doublet combinations (**Figure 4A**)
- However, triplet combination of OP-3136 with palazestrant and ribociclib increased inhibition compared to the doublet combination (**Figure 4B**)

Combination of OP-3136 with palazestrant leads to robust anti-tumor activity

Figure 5: Tumor growth inhibition (TGI) by combinations of KAT6 inhibitors with anti-estrogens and CDK4/6 inhibitors in a KAT6-overexpressing *ESR1*^{WT} ER+/HER2- breast cancer **cell line-derived xenograft (CDX) model**, **T47D** (scatter plot [**A**], tumor volume over time [**B**], waterfall plot [**C**])

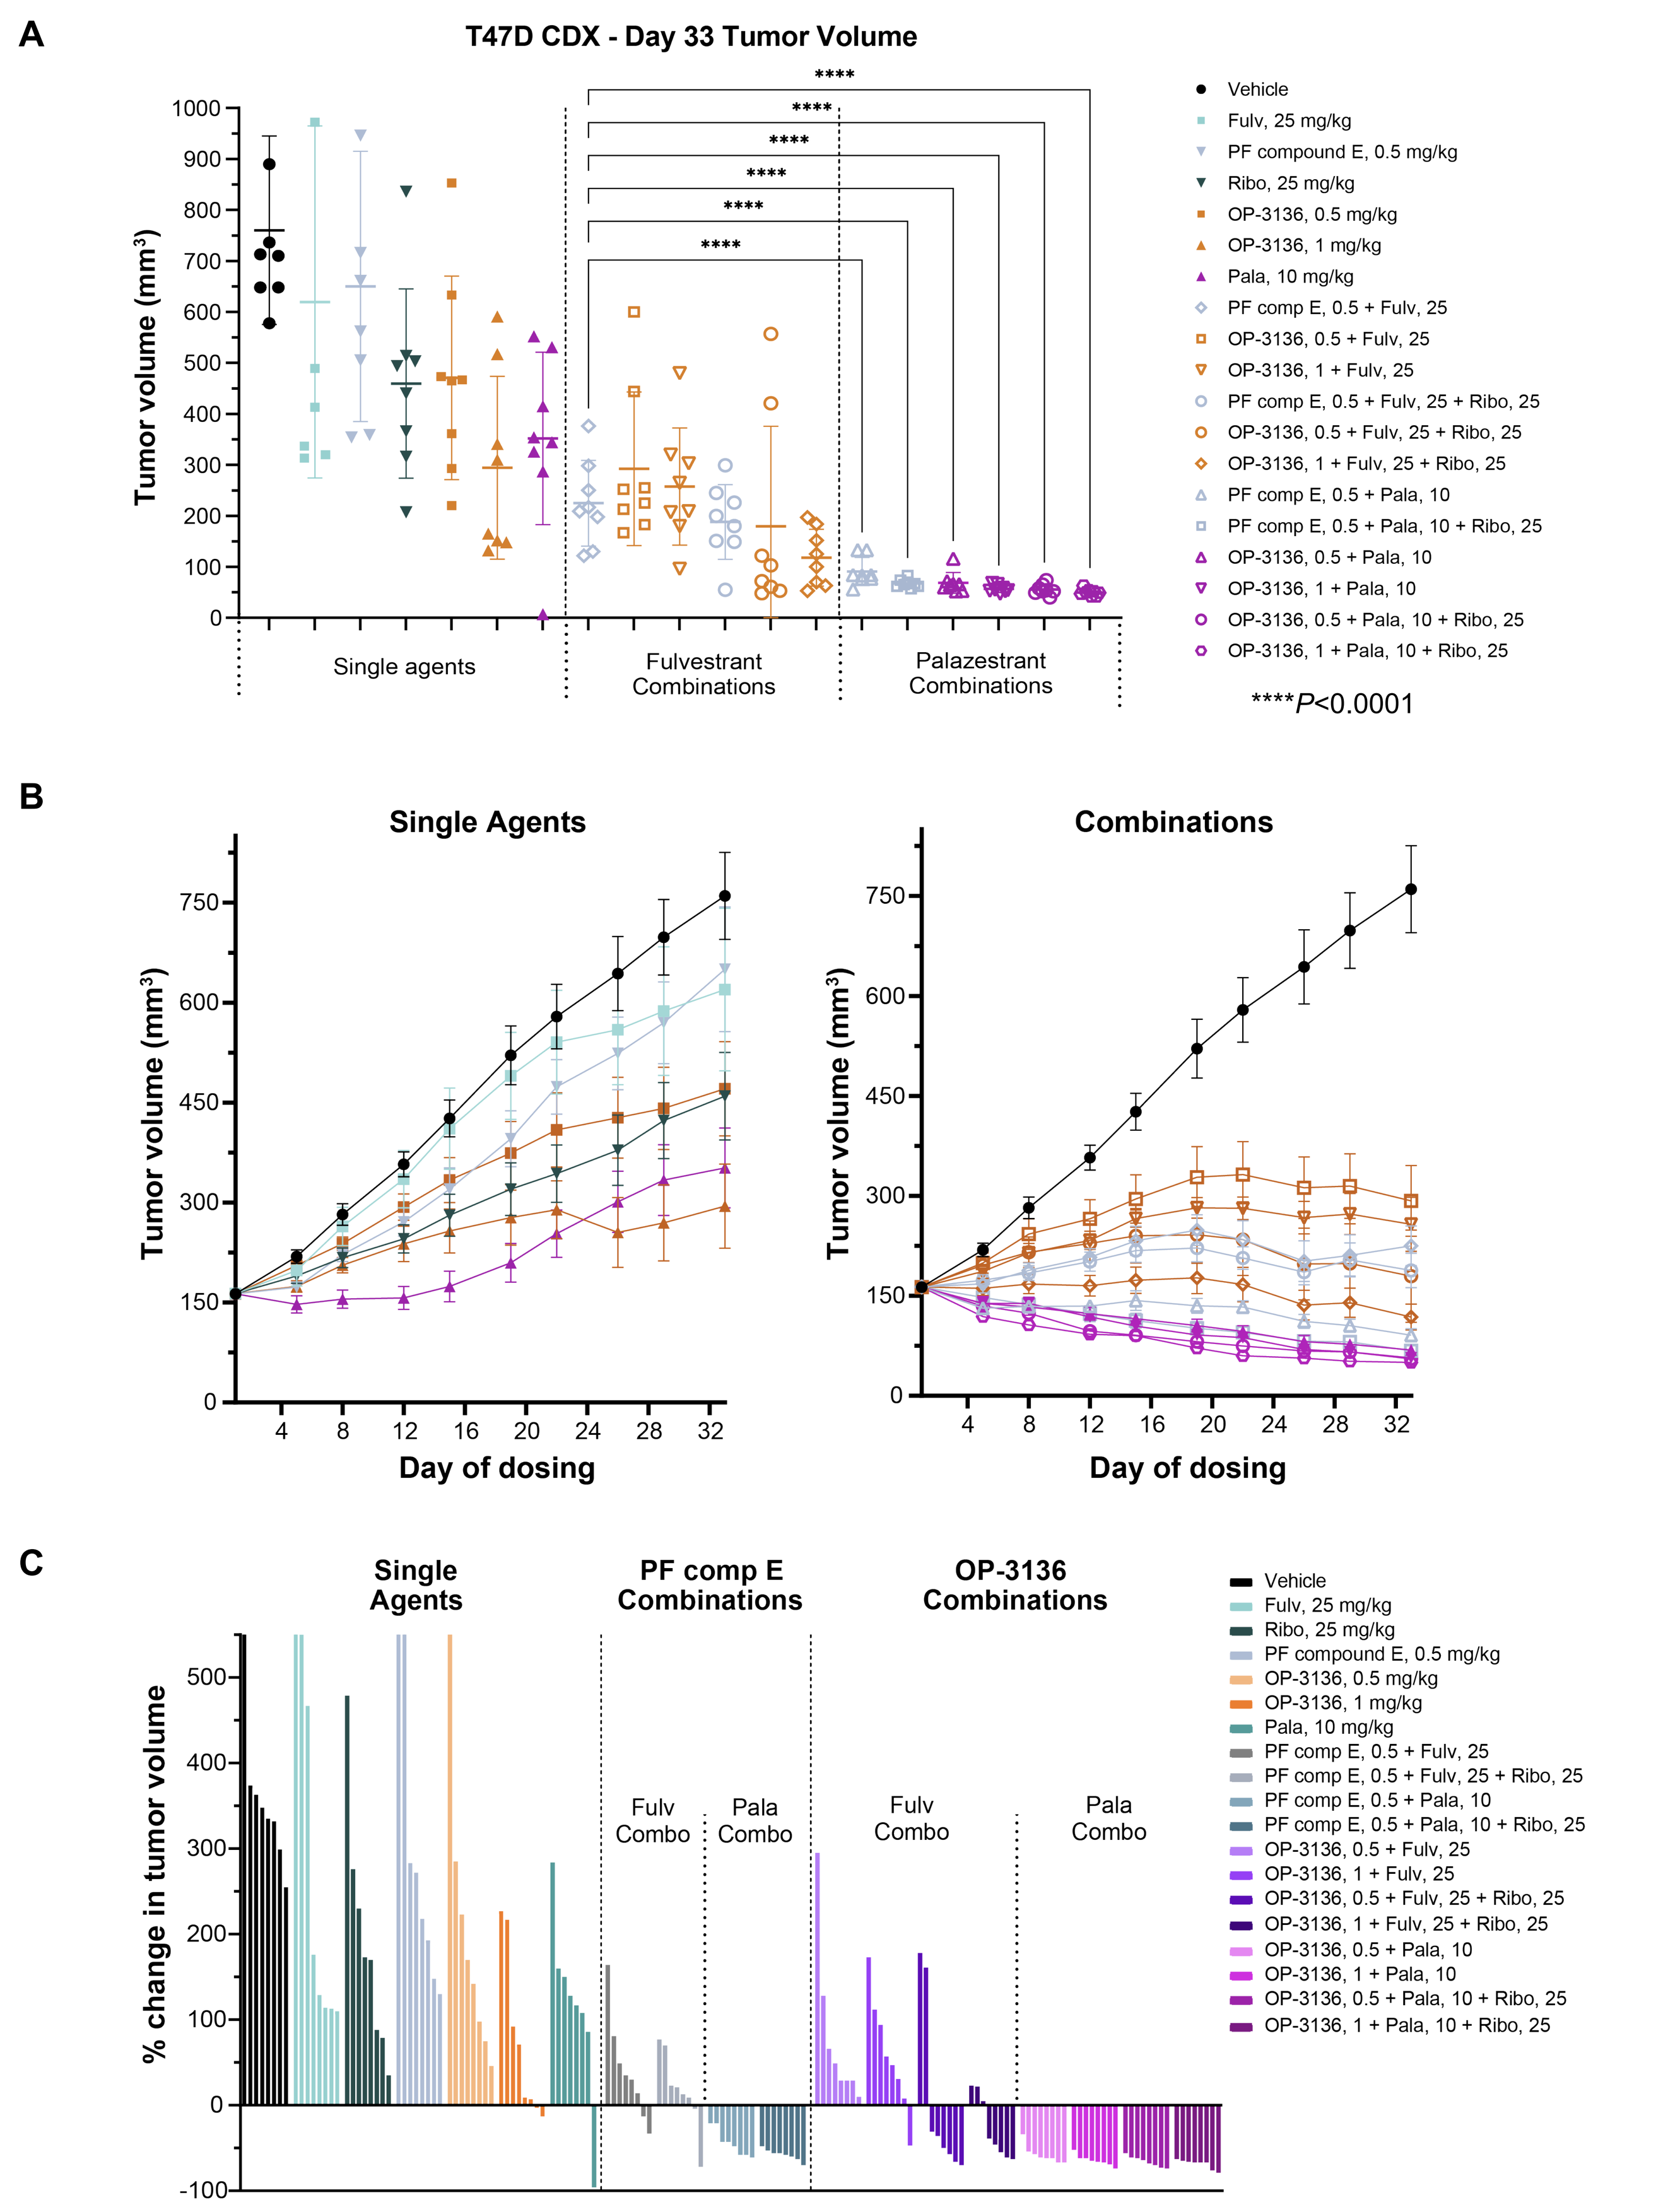
A
T47D CDX - Day 33 Tumor Volume
Tumor volume (mm³)
Single agents
Fulvestrant Combinations
Palazestrant Combinations
****P<0.0001
Vehicle
Fulv, 25 mg/kg
PF compound E, 0.5 mg/kg
Ribo, 25 mg/kg
OP-3136, 0.5 mg/kg
OP-3136, 1 mg/kg
Pala, 10 mg/kg
PF comp E, 0.5 + Fulv, 25
OP-3136, 0.5 + Fulv, 25
OP-3136, 1 + Fulv, 25
PF comp E, 0.5 + Fulv, 25 + Ribo, 25
OP-3136, 0.5 + Fulv, 25 + Ribo, 25
OP-3136, 1 + Fulv, 25 + Ribo, 25
PF comp E, 0.5 + Pala, 10
PF comp E, 0.5 + Pala, 10 + Ribo, 25
OP-3136, 0.5 + Pala, 10
OP-3136, 1 + Pala, 10
OP-3136, 0.5 + Pala, 10 + Ribo, 25
OP-3136, 1 + Pala, 10 + Ribo, 25
B
Single Agents
Combinations
Day of dosing
C
Single Agents
PF comp E Combinations
OP-3136 Combinations
Fulv Combo
Pala Combo
% change in tumor volume

Fulv, fulvestrant; **Pala**, palazestrant; **PF comp E,** PF compound E; **Ribo,** ribociclib

Figure 6: TGI by combinations of KAT6 inhibitors with anti-estrogens and CDK4/6 inhibitors in a KAT6-expressing *ESR1*^{WT} ER+/HER2+ **patient-derived xenograft (PDX) model, ST340** (scatter plot [**A**], tumor volume over time [**B**], waterfall plot [**C**])

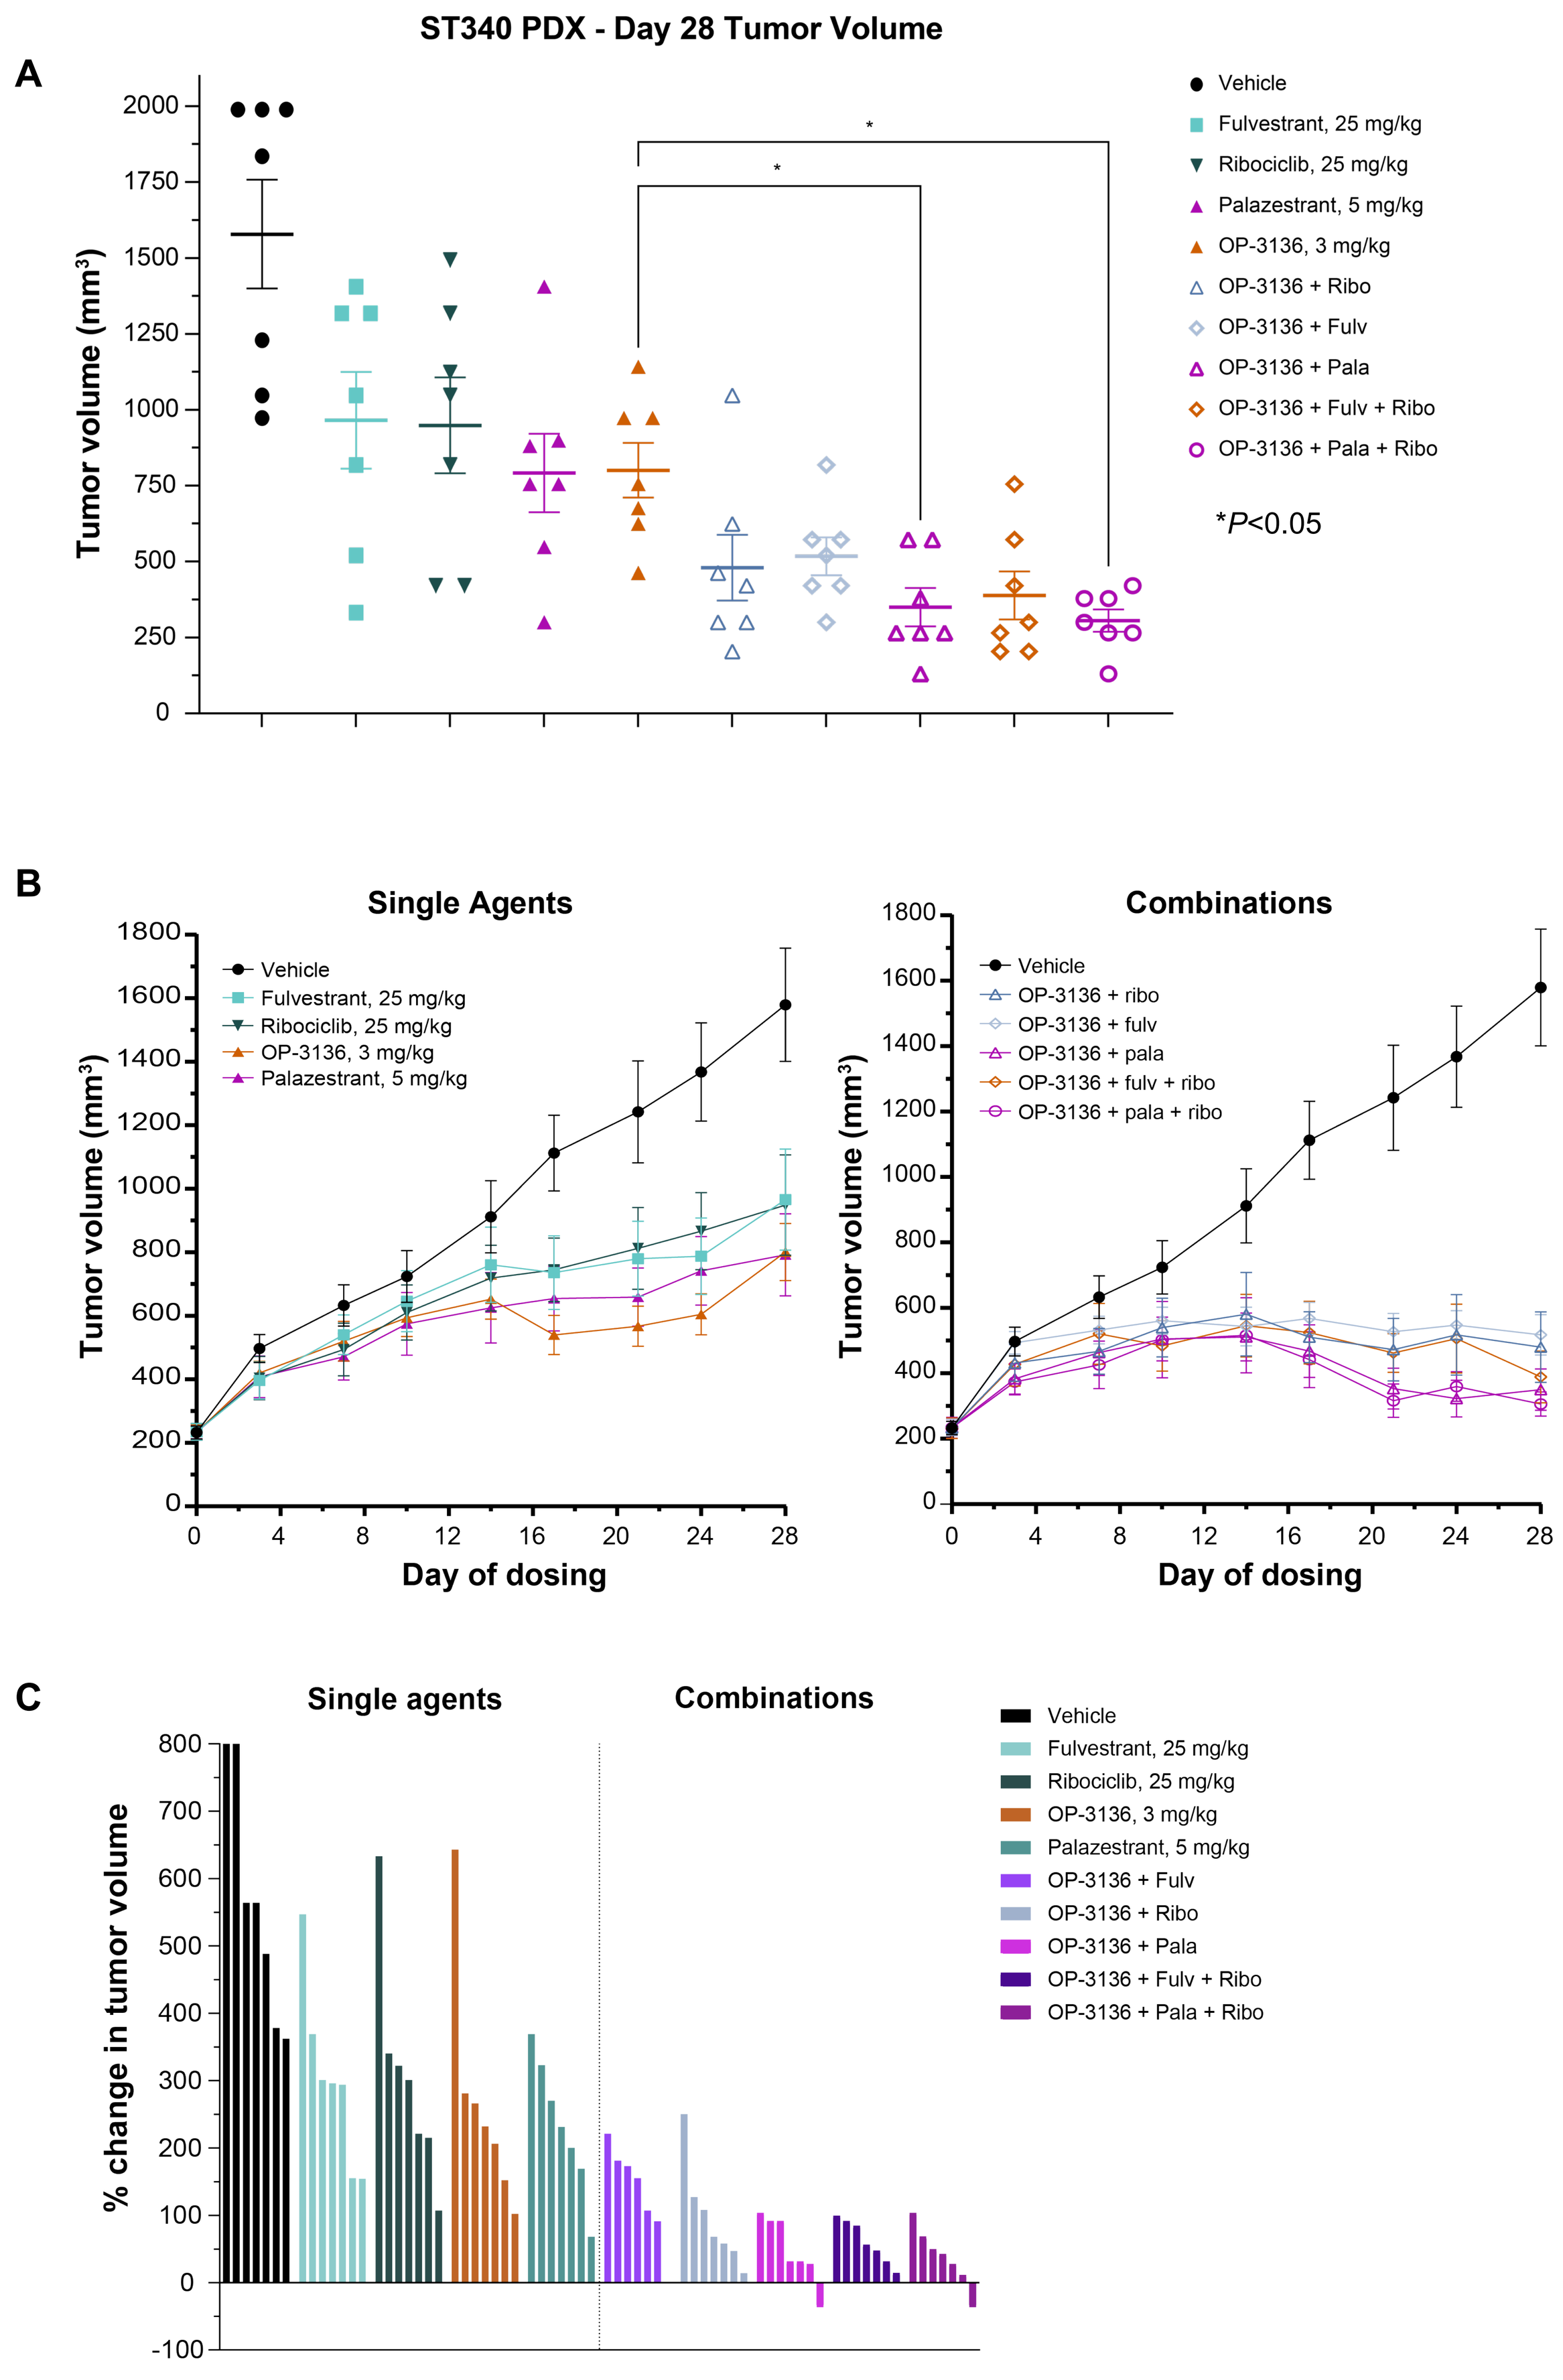
A
ST340 PDX - Day 28 Tumor Volume
Tumor volume (mm³)
Vehicle
Fulvestrant, 25 mg/kg
Ribociclib, 25 mg/kg
Palazestrant, 5 mg/kg
OP-3136, 3 mg/kg
OP-3136 + Ribo
OP-3136 + Fulv
OP-3136 + Pala
OP-3136 + Fulv + Ribo
OP-3136 + Pala + Ribo
*P<0.05
B
Single Agents
Combinations
Day of dosing
C
Single agents
Combinations
% change in tumor volume

Fulv, fulvestrant; **Pala,** palazestrant; **Ribo,** ribociclib

- OP-3136 at 1 mg/kg demonstrated the most robust TGI as monotherapy among all agents tested in a KAT6-overexpressing *ESR1*^{WT} ER+/HER2- breast cancer CDX model (**Figure 5A**)
- Combining fulvestrant with KAT6 inhibitors, OP-3136 or PF compound E, resulted in TGI, but not tumor regression, in a majority of the animals (**Figure 5B,C**)
- Combining palazestrant with OP-3136 or PF compound E resulted in significant tumor regression in all animals (**Figure 5B,C**)
- OP-3136 and palazestrant combinations showed significantly improved anti-tumor efficacy compared to PF compound E in combination with fulvestrant (**Figure 5C**)
- Addition of ribociclib to the combination of OP-3136 and palazestrant did not lead to improved anti-tumor response in T47D model (**Figure 5B,C**)
- In a KAT6-expressing *ESR1*^{WT} ER+/HER2+ PDX model, OP-3136 showed approximately 60% TGI as monotherapy (**Figure 6A,B**)
- Combination of OP-3136 and palazestrant with or without ribociclib showed significantly improved anti-tumor efficacy relative to the monotherapy (TGI of 91% and 95%, respectively) (**Figure 6B,C**)
- All OP-3136 combinations were well tolerated, with no significant changes in body weight and no mortality in either study

Conclusions

- **OP-3136 inhibited cell proliferation and synergized with anti-estrogens (fulvestrant and palazestrant) and a CDK4/6 inhibitor (ribociclib) in a breast cancer cell line.**
- **OP-3136 led to either tumor growth inhibition or tumor regression in vivo in T47D and ST340 xenograft models across all treatment groups.**
- **Palazestrant was consistently superior to fulvestrant when combined with KAT6 inhibitors and led to improved anti-tumor activity (tumor regression).**
- **OP-3136 showed robust synergistic anti-tumor activity when combined with either fulvestrant or palazestrant in doublet therapy, in both breast cancer CDX and PDX mouse models.**
- **The addition of ribociclib to the combination of OP-3136 and palazestrant (triplet therapy) provided no additional anti-tumor activity in the CDX and PDX models.**
- **OP-3136 is expected to enter a Phase 1 clinical trial in early 2025.**

References

1. Wiesel-Motiuk N, Assaraf YG. *Drug Resist Updat*. 2020;53:100729.
2. Sharma S et al. *Cell Chem Biol*. 2023;30(10):1191-1210.e20.
3. Yu L et al. *Oncogene*. 2017;36(20):2910-2918.
4. Mukohara T et al. *Nat Med*. 2024;30(8):2242-2250.
5. Palanisamy et al. Poster presented at: AACR-NCI-EORTC Annual Meeting; 2023; Boston, MA.
6. Paul AS et al. KAT6 inhibitor methods and combinations for cancer treatment. WO2022013369.

Poster presented at the 36th EORTC-NCI-AACR Symposium; 23-25 October 2024; Barcelona, Spain.

Acknowledgments

We thank our partner Aurigene for their role in the discovery of OP-3136.

This study was sponsored by Olema Oncology. Editorial and layout support were provided by Melanie Styers, PhD, and Shravanthi Mouli, PhD, of Verascity Science and funded by Olema Oncology.

For an e-Print, please scan the QR code. Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from the authors of this poster.

Sponsored by Olema Oncology